Modulating the tumor microenvironment via hypoxia activated prodrug CPD100Li induced cell death synergistically enhances anti-CTLA-4 immunotherapy

August Sick, Cascade Prodrug, Inc., Eugene, Oregon

Introduction:

The industry is diverting from chemotherapy because of side-effects, primarily immunosuppressive – but not so fast, it could hold immunostimulatory properties. CPD100, a prodrug, is activated in the hypoxic regions of the tumor microenvironment [1, 2]. The liposome formulation, CPD100Li, targets the prodrug to the tumor following the enhanced permeability and retention effect (EPR). Our hypothesis suggests that indiscriminate cell death induced in the hypoxic regions of solid tumors by activating a potent cytotoxin could effectively remodel the tumor microenvironment from tumor directed immunosuppression to modulated immunity under the control of one or more checkpoint molecules. Combining CPD100Li therapy with a checkpoint inhibitor could then unleash activated T cells to kill the cancer selectively.

Materials and Methods:

1. Immunophenotype Study – CPD100Li
 - Female Balb/c mice (from Envigo) were used for subcutaneous tumor implants of CT26 cells in the right high axilla. Tumor growth was monitored by caliper measurements.
 - Mice were staged when tumors reached 100mm3

Group	N	Treatment	Dose	ROA	Regimen	Days of Treatment
1	6	Vehicle	0.21 ml/20 g	IV	Q7Dx2	8, 15
2	6	Isotype control	40 mg/kg	IV	Q7Dx2	8, 15

 - All animals were euthanized by cervical dislocation to prevent introduction of a hypoxic environment.
 - For tumor immune profiling, tumors from all mice were collected 24 hours post last dose, digested to a single cell suspension for flow cytometry (Miltenyi, Germany). CompLeukocyteTM panels were used on an Attune NxT Flow Cytometer (Thermo Fisher Scientific) and analyzed with FlowJo software (Tree Star, Inc. Ashland, OR).

2. Efficacy Study - Combination CPD100Li and Immune Checkpoint Inhibitors
 - Female Balb/c mice (from Envigo) were used for subcutaneous tumor implants of CT26 cells in the right high axilla. Tumor growth was monitored by caliper measurements.
 - Mice were staged into groups of 8 when tumors reached 100mm3.

Group	Treatment	Dose	ROA	Regimen	Days of Treatment
1	Vehicle	0.16 ml/20 g	IV	Q14Dx2	7, 21
	Isotype control	10 mg/kg	IP	(Q3Dx2,10)(x2)	7, 10, 21, 24
2	CPD100Li	30 mg/kg	IV	Q14Dx3	7, 21, 35
3	CPD100Li	30 mg/kg	IV	Q14Dx3	7, 21, 35
	Isotype control	10 mg/kg	IP	(Q3Dx2,10)(x2);QDx1(D35)	7, 10, 21, 24, 35
4	Anti-mCTLA-4	10 mg/kg	IP	(Q3Dx2,10)(x2)	7, 10, 21, 24
5	Anti-mPD-L1	10 mg/kg	IP	Q3Dx2; QDx1(D21)	7, 10, 21
6	Anti-mVISTA	10 mg/kg	IP	Q3Dx2; QDx1(D21)	7, 10, 21
7	CPD100Li	30 mg/kg	IV	Q14Dx3	7, 21, 35
	Anti-CTLA-4	10 mg/kg	IP	(Q3Dx2,10)(x2);QDx1(D35)	7, 10, 21, 24, 35
8	CPD100Li	30 mg/kg	IV	Q14Dx2	7, 21
	Anti-mPD-L1	10 mg/kg	IP	Q3Dx2; QDx1(D21)	7, 10, 21
9	CPD100Li	30 mg/kg	IV	Q14Dx2	7, 21
	Anti-mVISTA	10 mg/kg	IP	Q3Dx2; QDx1(D21)	7, 10, 21

 - Checkpoint inhibitor antibodies were from BioXCell (West Lebanon, NH); Isotype control Clone MPC-11, anti-mCTLA-4 Clone 9D9, anti-mPD-L1 Clone 10F.9G2, anti-VISTA Clone 13F3.
 - Disease progression was monitored by Tumor, Body Weight measurements 3x/week, Increased Time to Progression (ITP) Complete Regression (CR) and Tumor Free Survivors (TFS

Results:

Flow cytometry demonstrated CPD100Li decreased cell viability in the tumors compared to Vehicle and triggered an increase in the percentage of CD45+ among total live cells (Figure 1). CPD100Li treatment also led to decreased absolute cell counts of Tregs, M-MDSC and M2 macrophages, compared to Vehicle but maintained the percentage of M1 macrophages (Figure 2). Furthermore, the ratio of M1/M2 tumor-associated macrophages increased, suggesting that CPD100Li enhances the persistence of immune cells in the tumor (Figure 3). Finally, a reduction in the expression of PD-1 exhaustion marker on CD8+ T cells was observed (Figure 4). None of the checkpoint inhibitors alone demonstrated a strong response to delaying tumor growth whereas the combination of CPD100Li and CTLA-4 showed a significant synergistic response delaying tumor growth – an increase in time to progression of >200%, and a >60% incidence of complete tumor regressions with 25% remaining as tumor-free survivors at the end of the study (Figure 5).

Conclusions:

Taken together, these data provide evidence that CPD100Li inhibits CT26 tumor growth by attenuating T cell exhaustion, possibly by a mechanism that promotes M1 macrophage activity. Feasibly, that CPD100Li induces changes in the tumor microenvironment that can convert “cold” tumors into “hot”, promoting a strong anti-tumor response in combination with CTLA-4. This study demonstrates a novel and effective combination strategy for cancer immunotherapy, making CPD100Li a potent candidate for cancer therapy in the near future.

Figure 1. A) Mean body weight, B) Mean Tumor volume, C) Live cells (Zombie Viability Dye) as a percentage of total cells and D) CD45 positive cells as a percentage of live cells

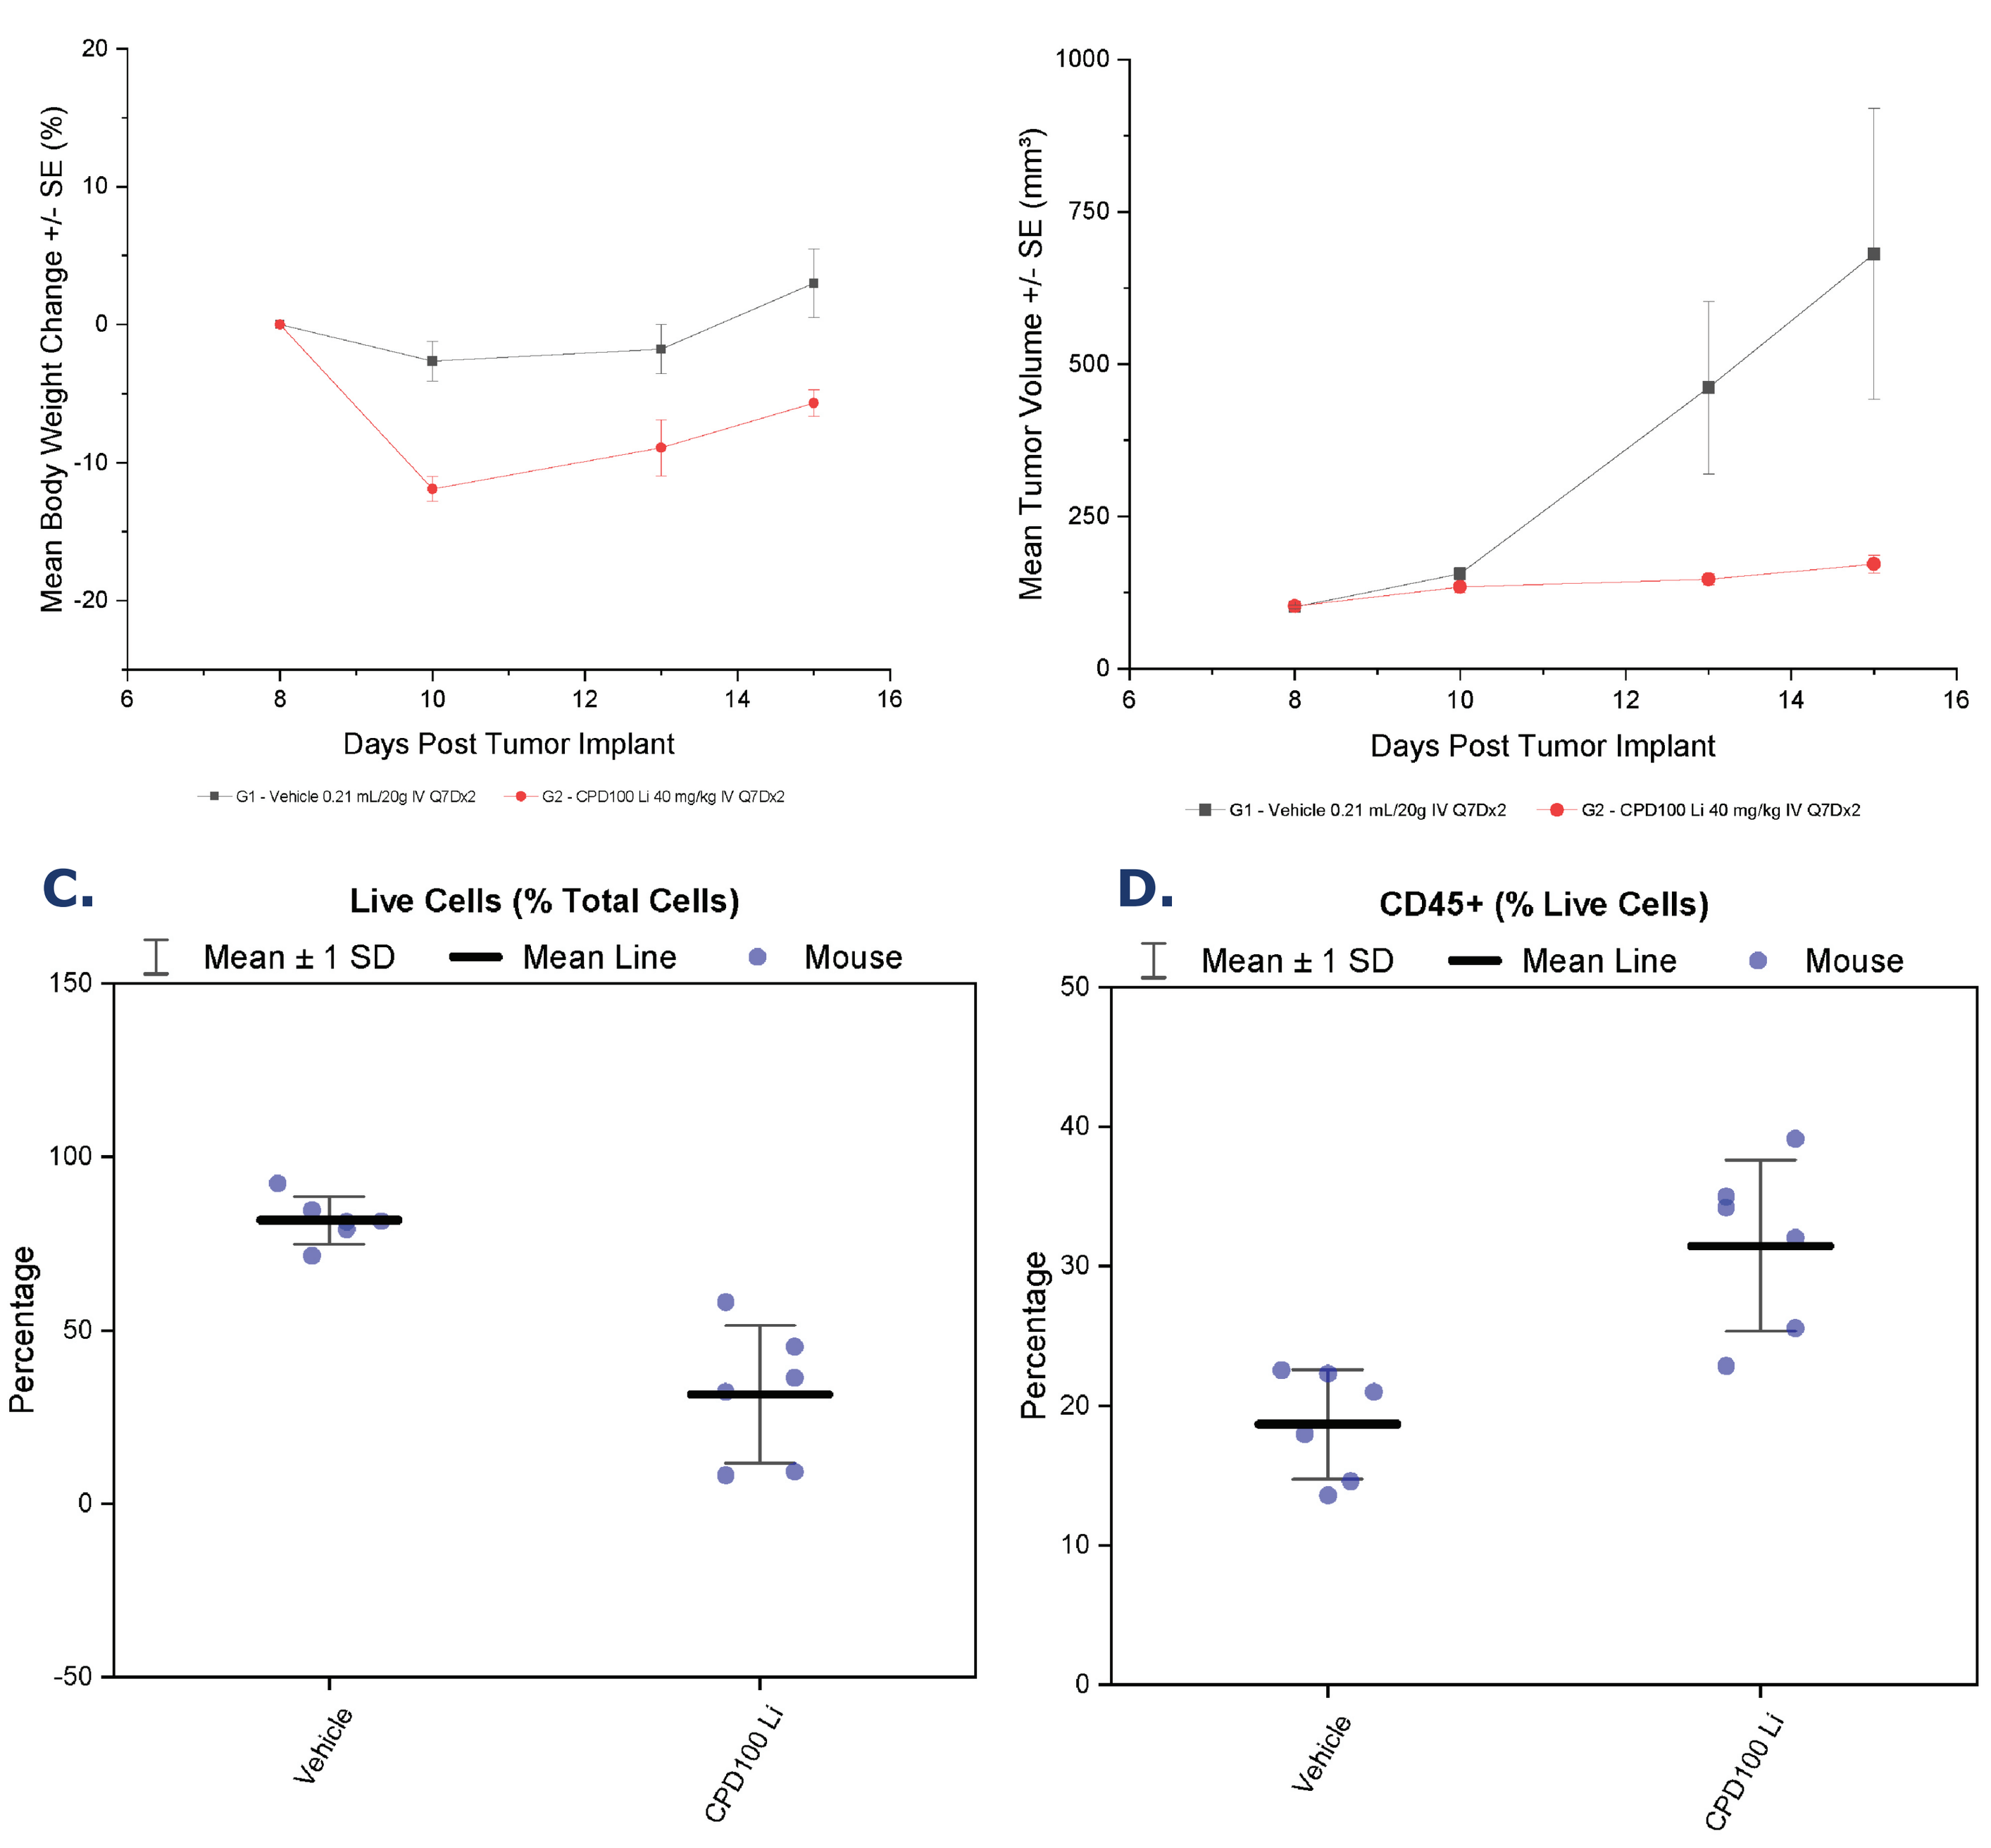
A.
Mean Body Weight Change +/- SE (%)
Days Post Tumor Implant
G1 - Vehicle 0.21 mL/20g IV Q7Dx2
G2 - CPD100 Li 40 mg/kg IV Q7Dx2
B.
Mean Tumor Volume +/- SE (mm³)
Days Post Tumor Implant
G1 - Vehicle 0.21 mL/20g IV Q7Dx2
G2 - CPD100 Li 40 mg/kg IV Q7Dx2
C.
Live Cells (% Total Cells)
Mean ± 1 SD
Mean Line
Mouse
Percentage
Vehicle
CPD100 Li
D.
CD45+ (% Live Cells)
Mean ± 1 SD
Mean Line
Mouse
Percentage
Vehicle
CPD100 Li

Figure 2. Absolute cell counts A) Treg, B) M2 Macrophage and C) M-MDSC

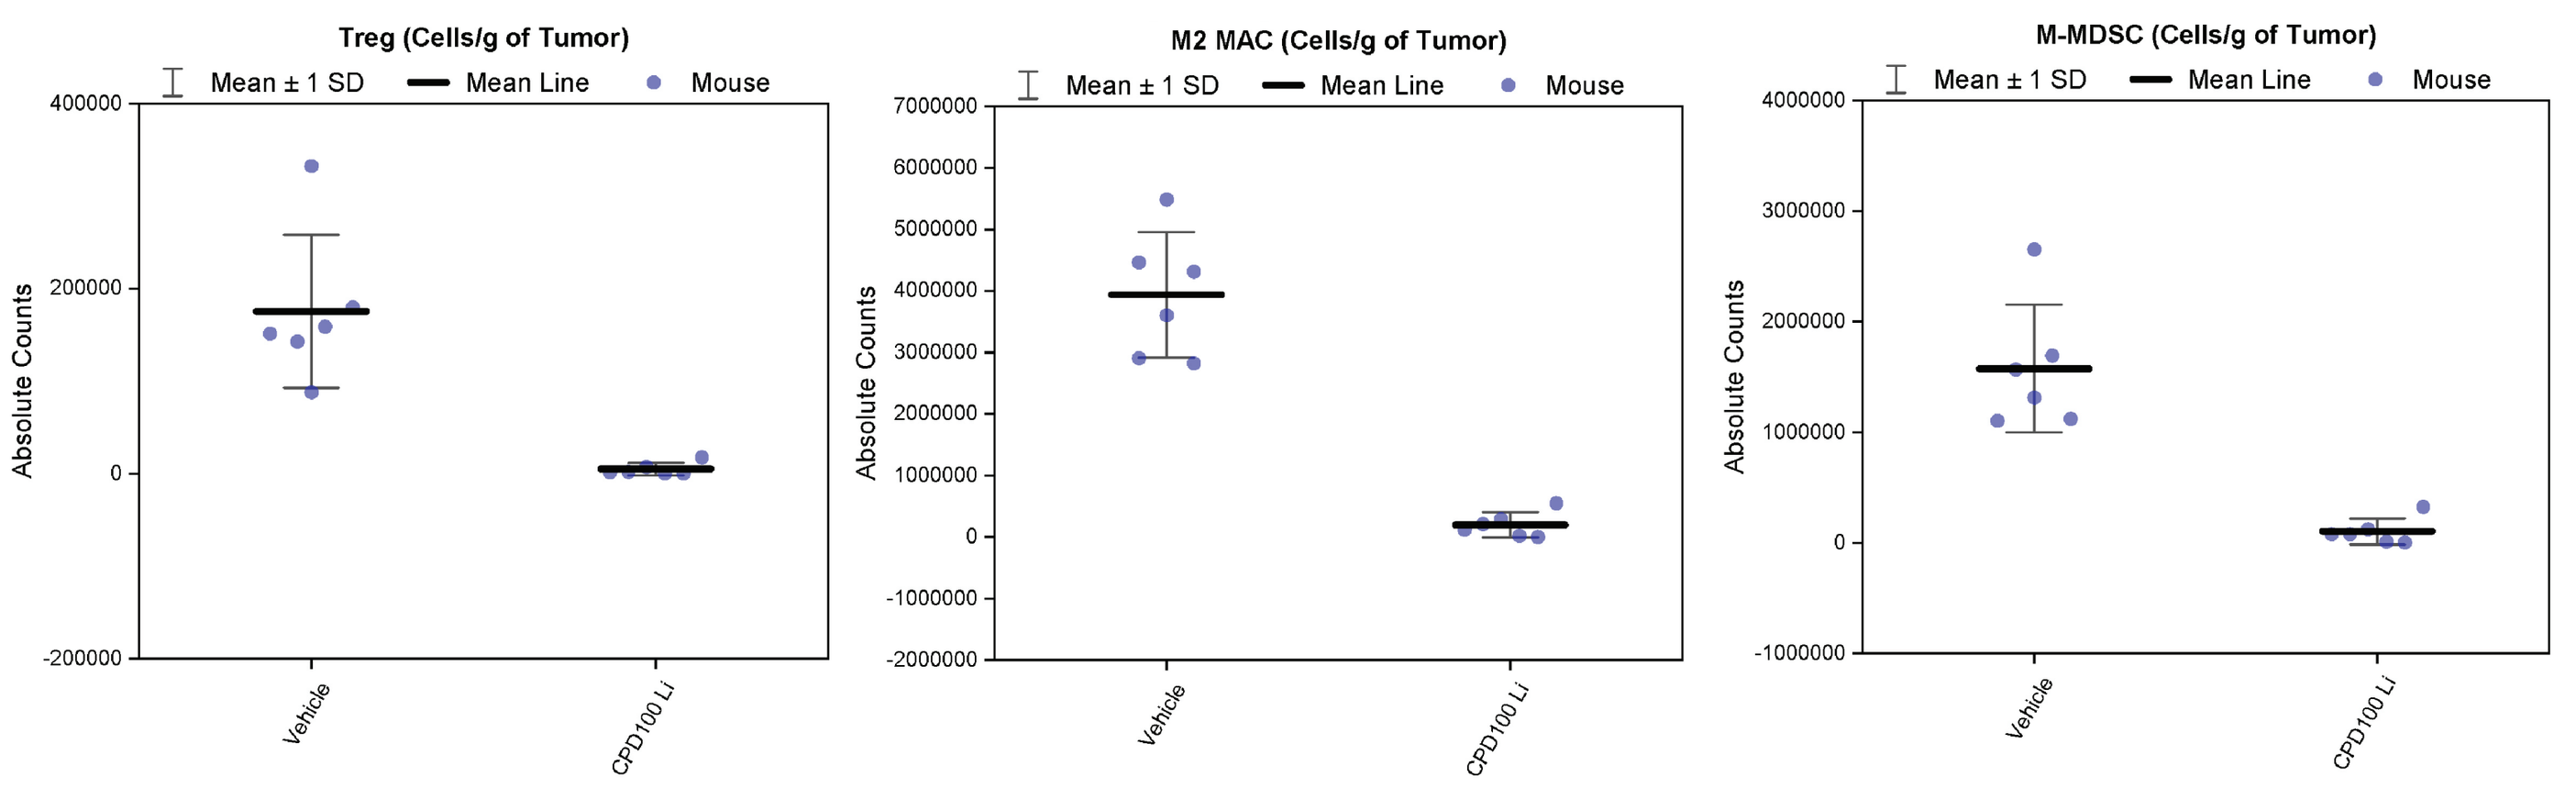
A.
Treg (Cells/g of Tumor)
B.
M2 MAC (Cells/g of Tumor)
C.
M-MDSC (Cells/g of Tumor)
Mean ± 1 SD
Mean Line
Mouse
Absolute Counts
Vehicle
CPD100 Li

Figure 3. Ratio M1/M2 Macrophage absolute cell counts

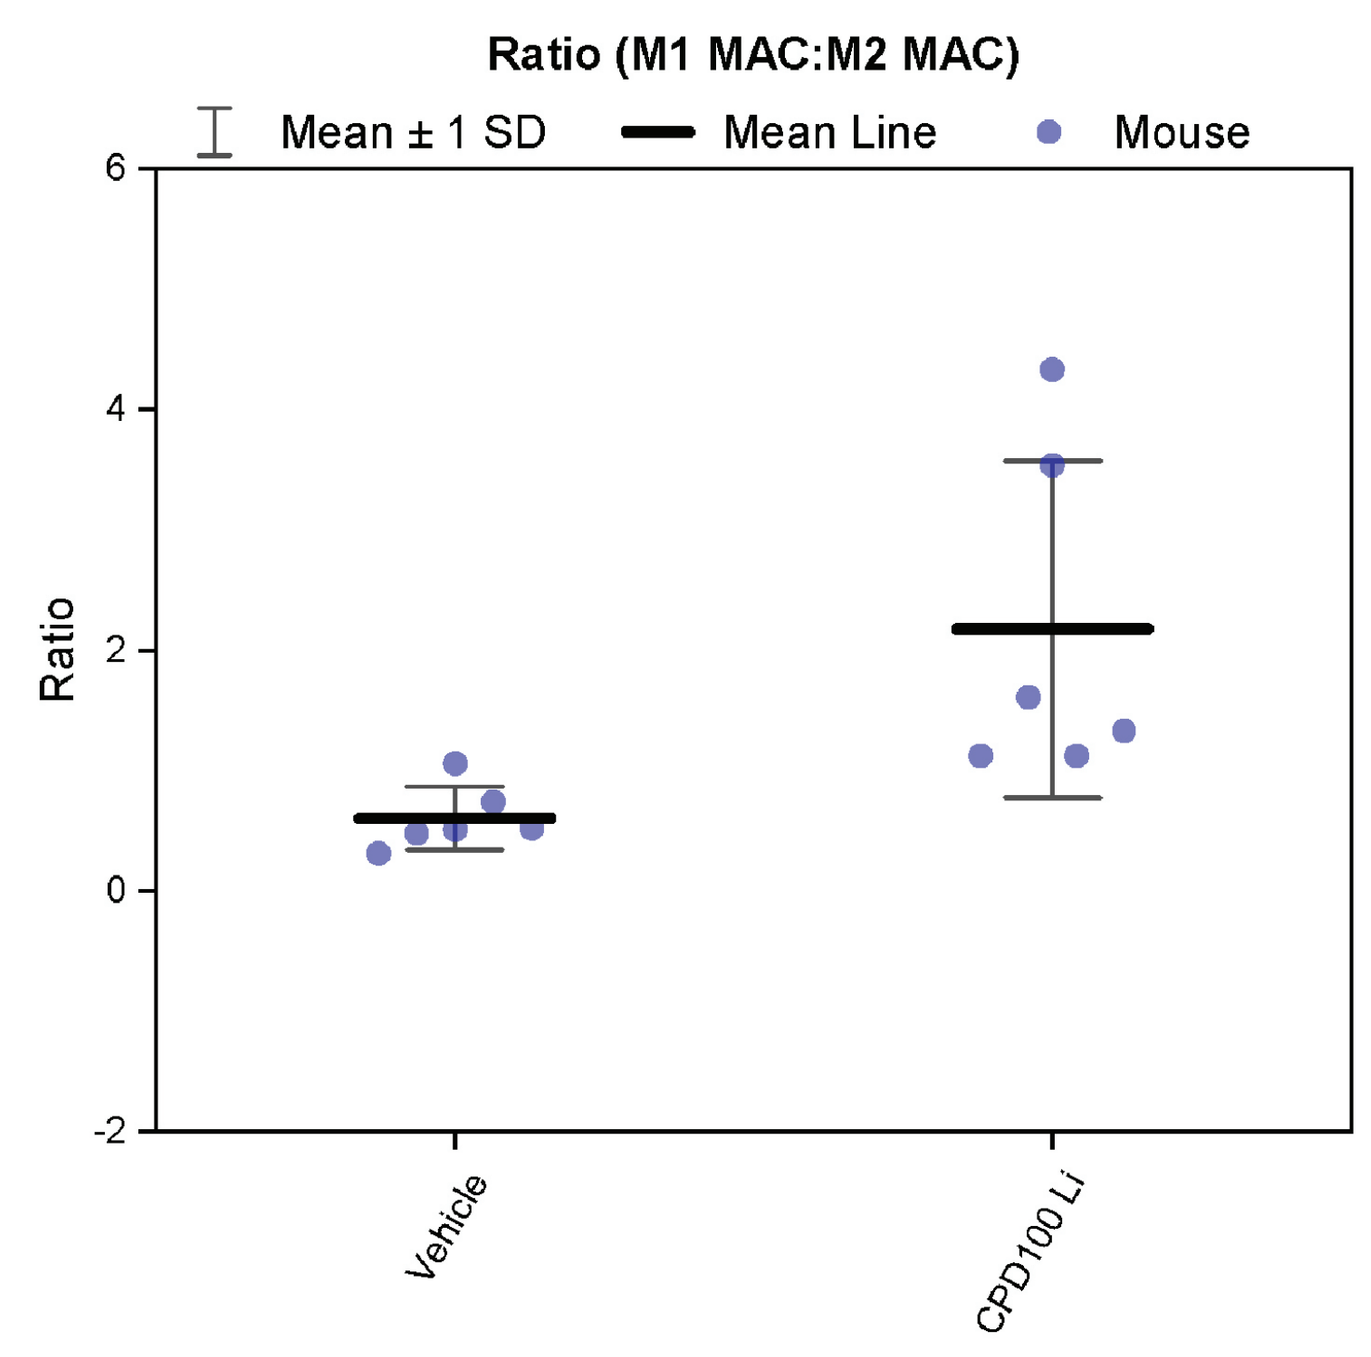
Ratio (M1 MAC:M2 MAC)
Mean ± 1 SD
Mean Line
Mouse
Ratio
Vehicle
CPD100 Li

Figure 4. PD-1+ cells as a percentage of CD8+ T cells

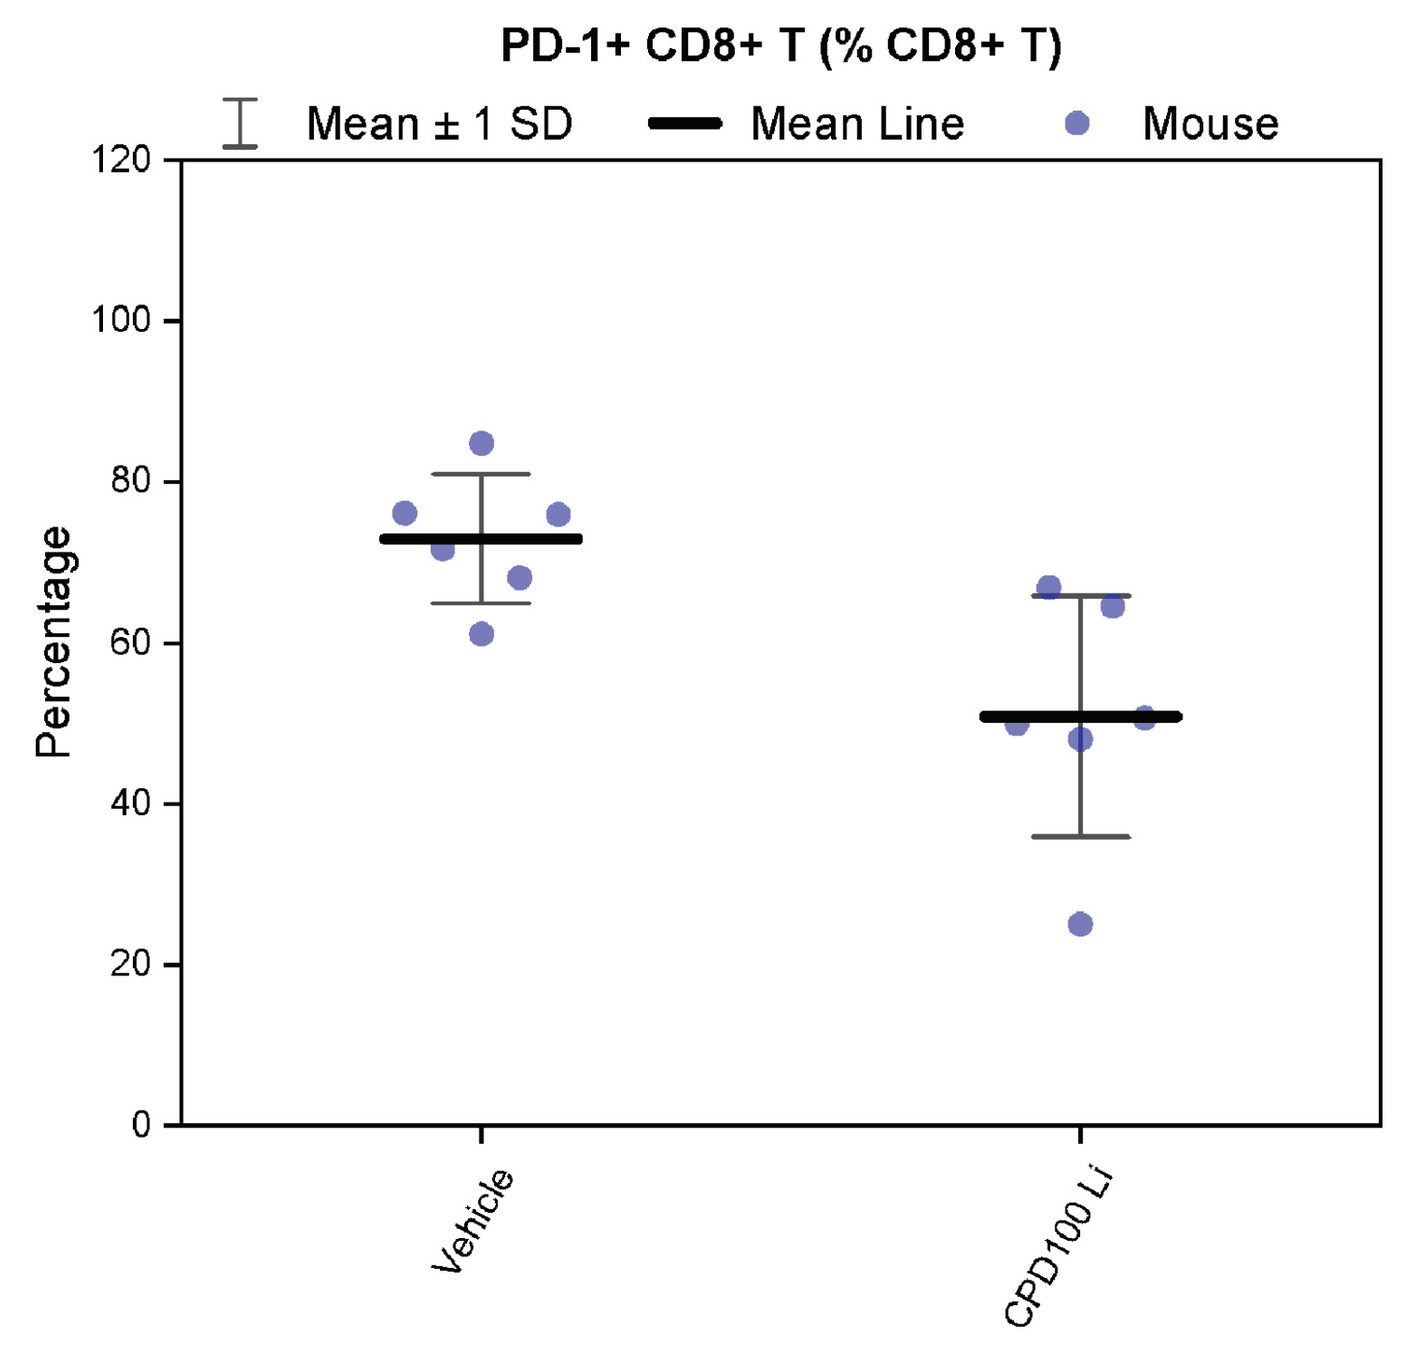
PD-1+ CD8+ T (% CD8+ T)
Mean ± 1 SD
Mean Line
Mouse
Percentage
Vehicle
CPD100 Li

Figure 5. A) Body weight, B) Tumor growth, C) Disease Progression Endpoints; Increased Time to Progression (ITP), Complete Regression (CR), Tumor Free Survivors (TFS)

A.

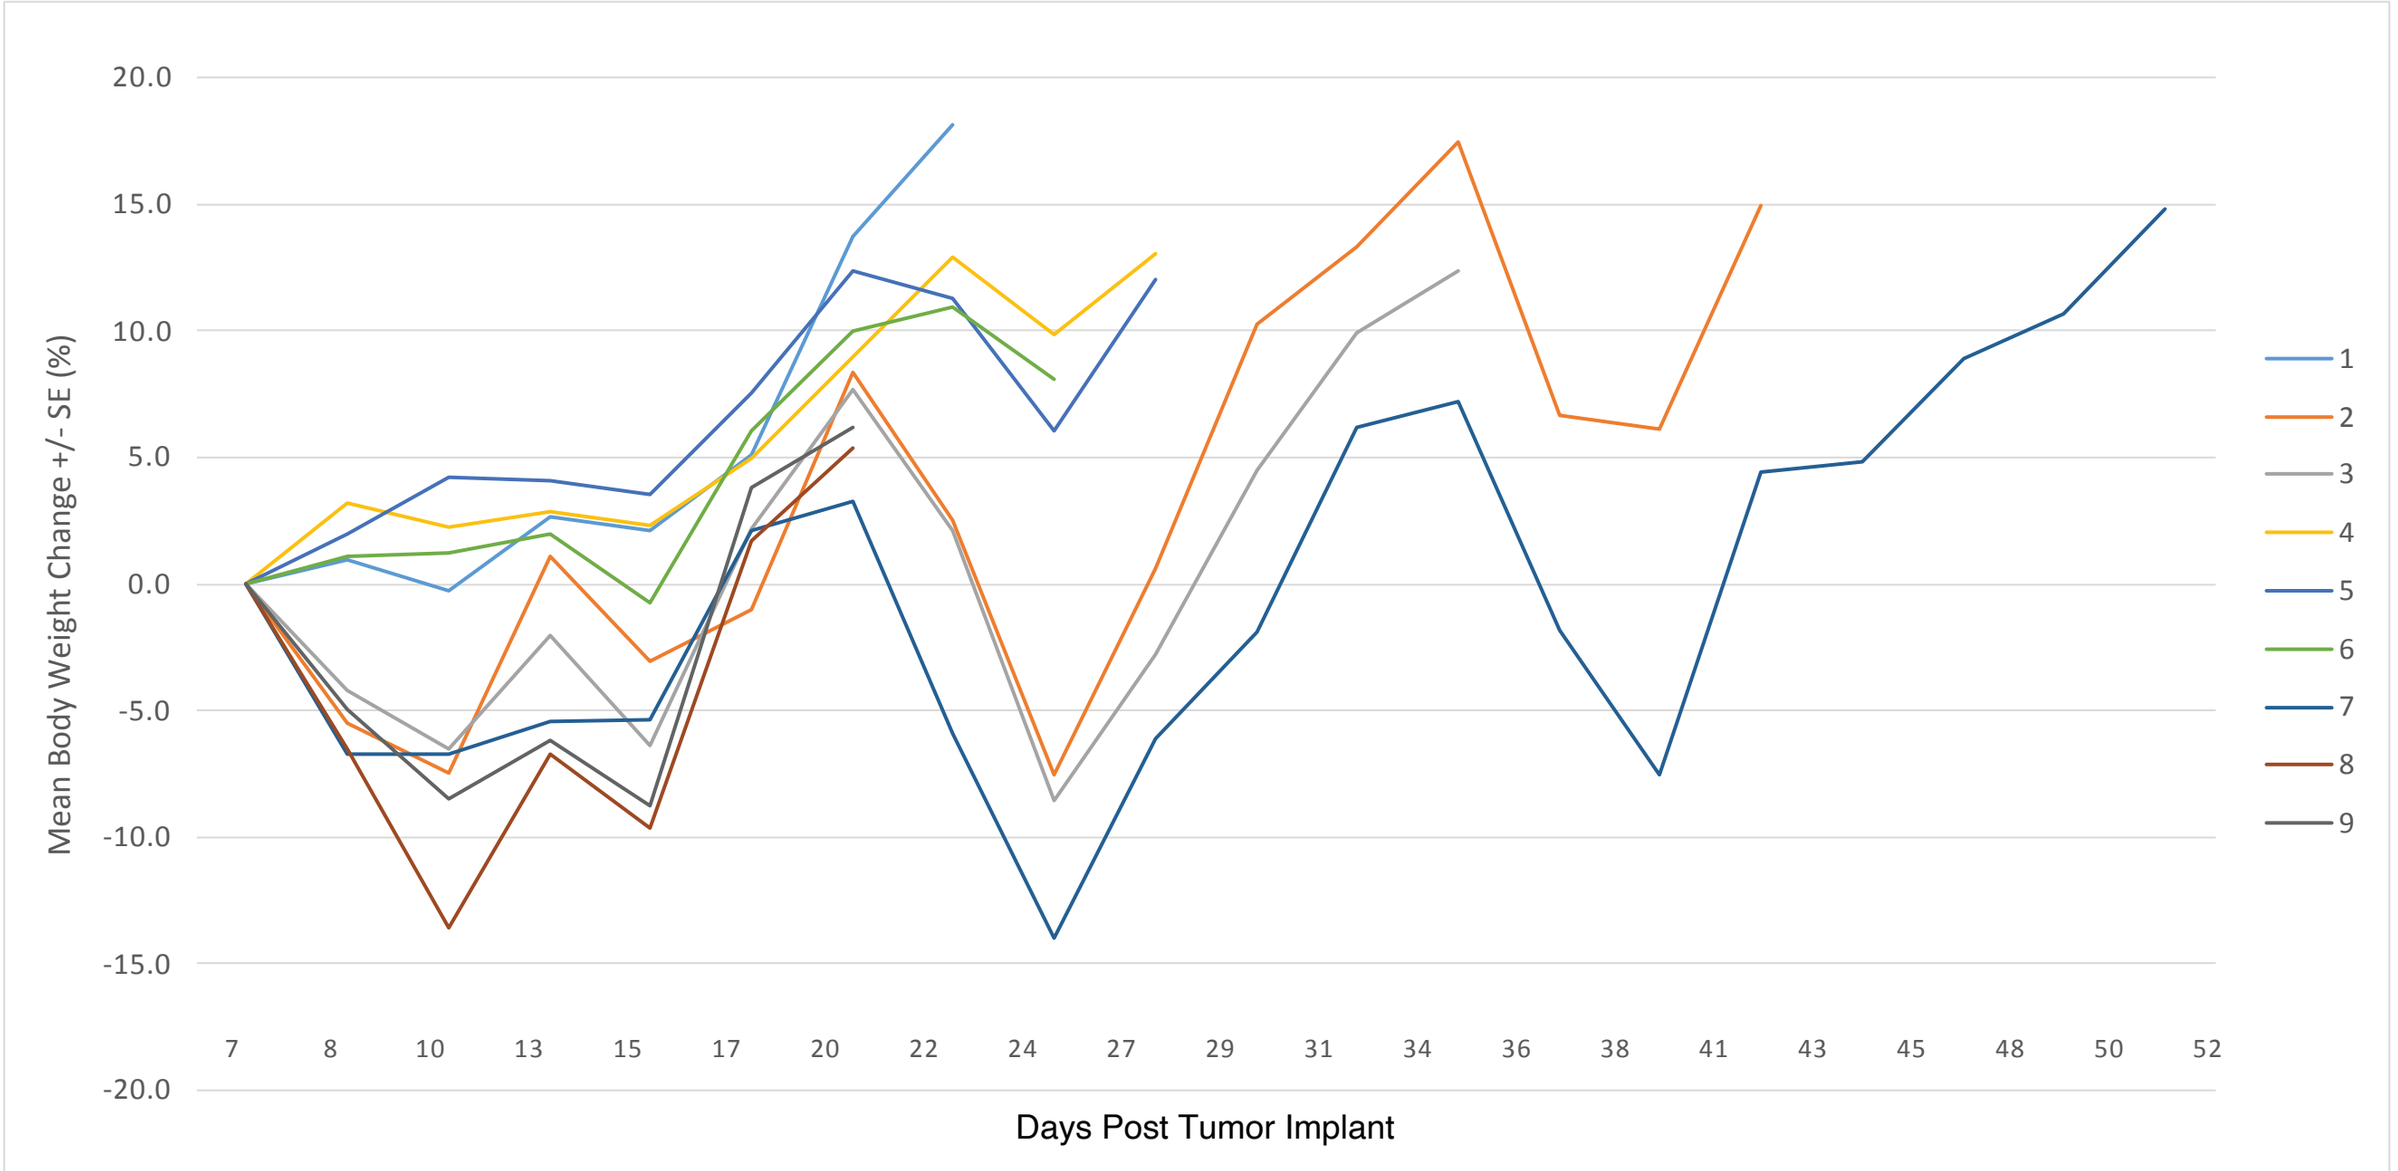
Mean Body Weight Change +/- SE (%)
Days Post Tumor Implant

B.

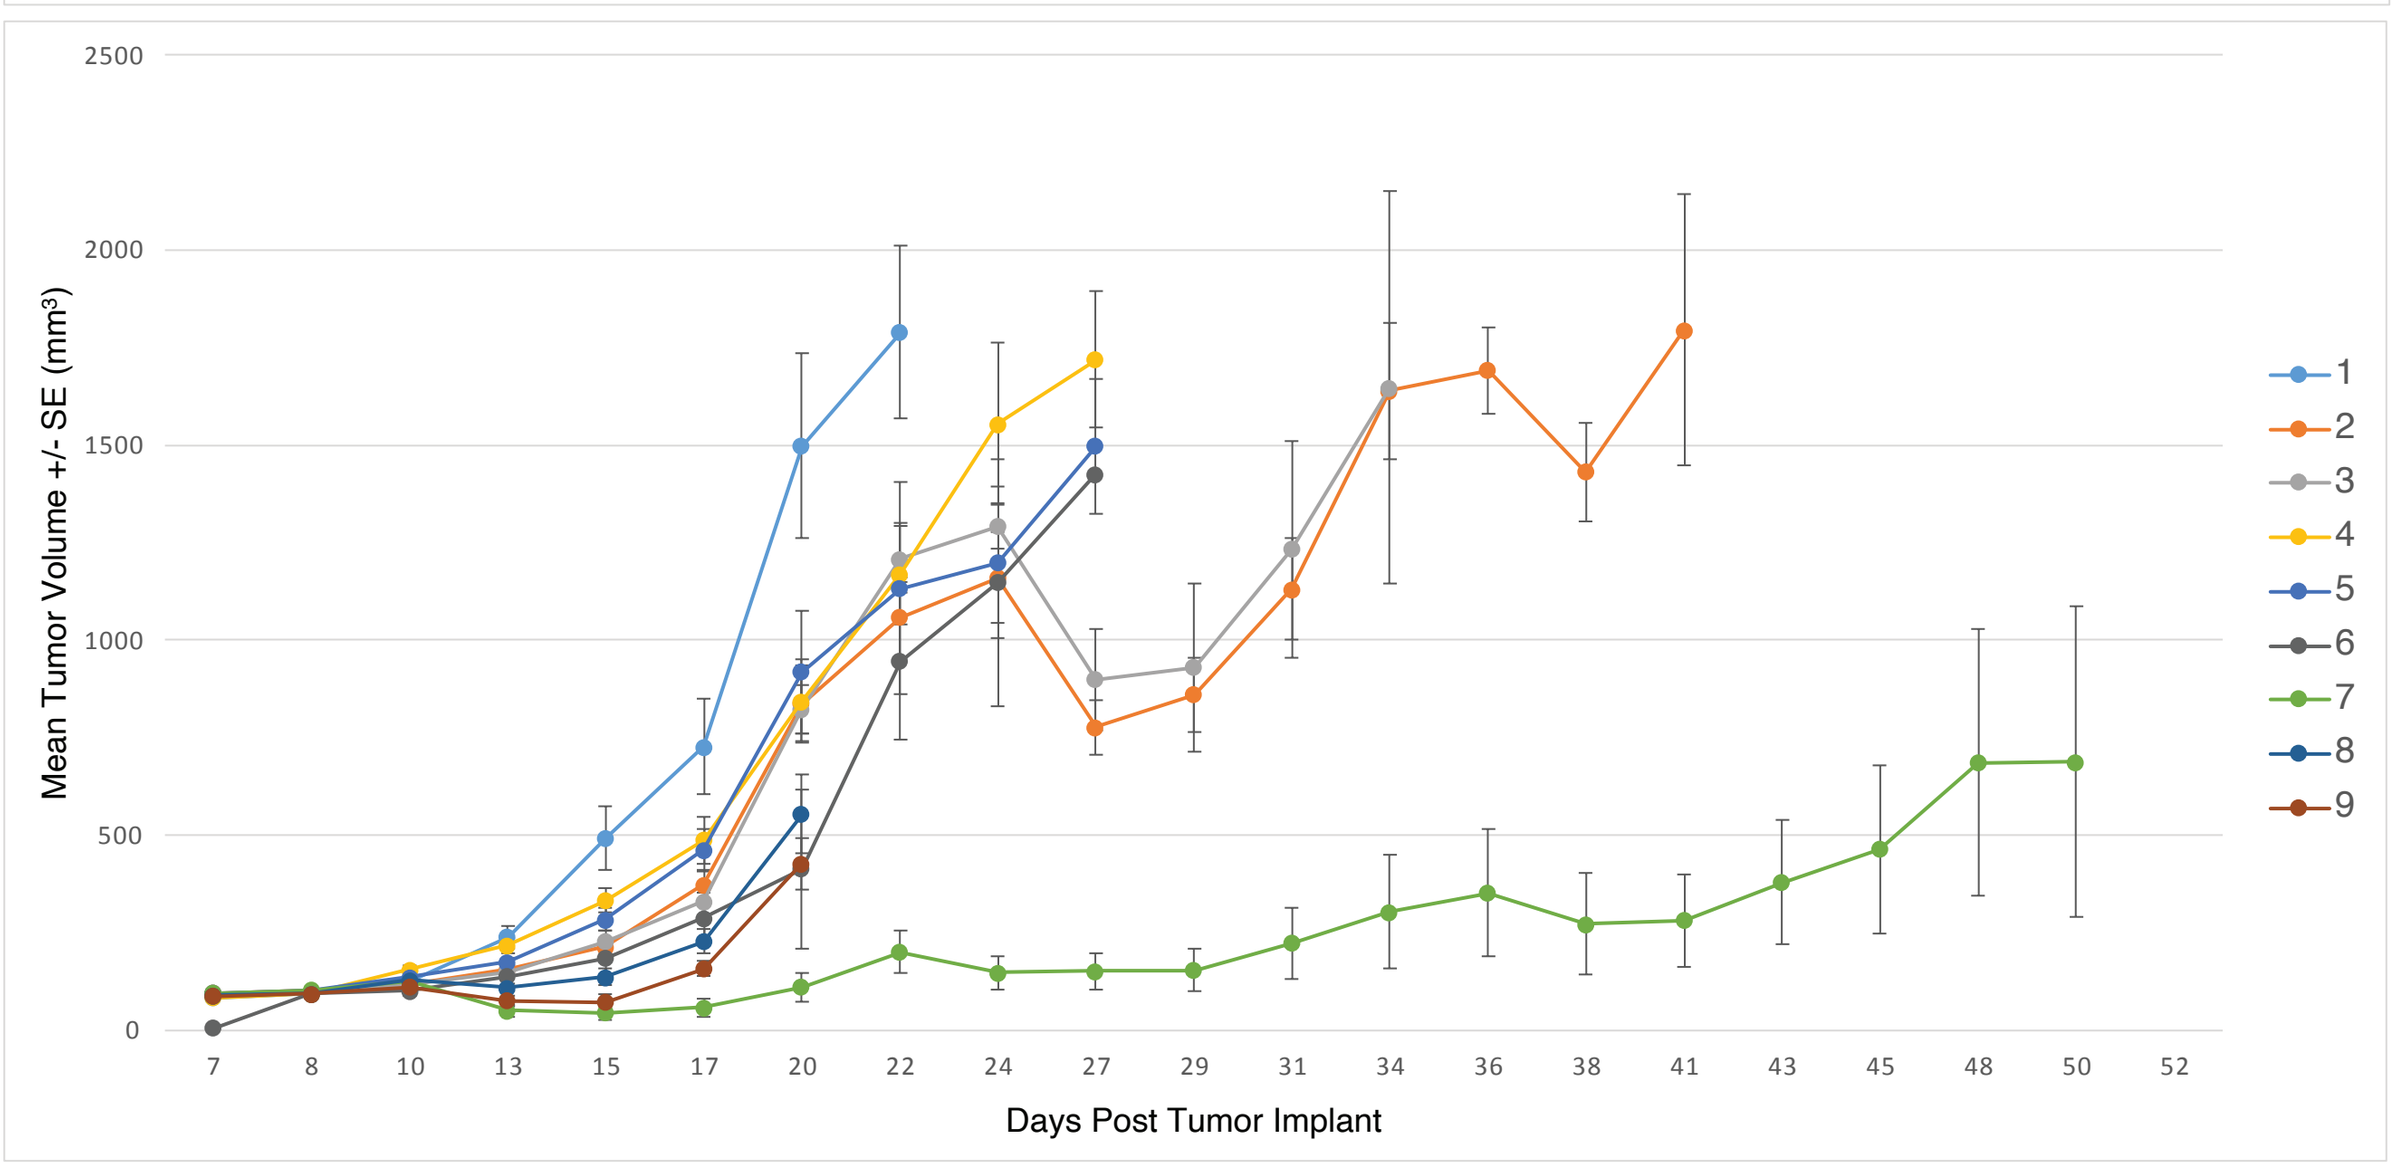
Mean Tumor Volume +/- SE (mm³)
Days Post Tumor Implant

C.

Group	Treatment	Increase Time to Progression (%)	Complete Regression (%)	Tumor Free Survivor (%)
1	Vehicle + Isotype Control	NA	0.0	0.0
2	CPD100Li	128	0.0	0.0
3	CPD100Li + Isotype Control	71	0.0	0.0
4	Anti-mCTLA-4	35	0.0	0.0
5	Anti-mPD-L1	42	0.0	0.0
6	Anti-mVISTA	21	0.0	0.0
7	CPD100Li + anti-mCTLA-4	>207	62.5	25.0
8	CPD100Li + anti-mPD-L1	7	0.0	0.0
9	CPD100Li + anti-mVISTA	7	0.0	0.0

References
Vidhi M. Shah, Duc X. Nguyen, Adel Alfatease, Shay Bracha, Adam WG. Alani, Characterization of pegylated and non-pegylated liposomal formulation for the delivery of hypoxia activated vinblastine-N-oxide for the treatment of solid tumors, Journal of Controlled Release, Volume 253, 2017, Pages 37-45,
Shah, V.M.; Dorrell, C.; Al-Fatease, A.; Allen-Petersen, B.L.; Woo, Y.; Bortnyak, Y.; Gheewala, R.; Sheppard, B.C.; Sears, R.C.; Alani, A.W. Microfluidics Formulated Liposomes of Hypoxia Activated Prodrug for Treatment of Pancreatic Cancer. Pharmaceutics 2022, 14, 713.

Studies were performed by Labcorp Drug Development. All animal work was performed in an AAALAC accredited facility, in alignment with applicable animal welfare regulations.

Presented at the 36th Annual Meeting of the Society for Immunotherapy of Cancer (SITC), November 3 - 5, 2023, San Diego, CA
© 2023 Cascade Prodrug, Inc.

Cascade PRODRUG™

www.cascadeprodrug.com